High-Throughput Single Molecule Tracking Reveals Estrogen Receptor Protein Interaction Landscape in Living Cells

David T. McSwiggen, Helen Liu, Ruensern Tan, Sebastia Agramunt Puig, Lakshmi B. Akella, Russell Berman, Mason Bretan, Hanzhe Chen, Xavier Darzacq, Kelsey Ford, Ruth Godbey, Eric Gonzalez, Adi Hanuka, Alec Heckert, Jaclyn J. Ho, Stephanie L. Johnson, Reed Kelso, Aaron Klammer, Jifu Li, Kevin Lin, Brian Margolin, Patrick McNamara, Laurence Meyer, Sarah E. Pierce, Akshay Sule, Yangzhong Tang, Daniel J. Anderson, Hilary P. Beck

Eikon Therapeutics, 3929 Point Eden Way, Hayward CA, 94545

Abstract

Protein motion is defined by a network of interactions within the cellular environment, serving as a direct report of activity and function. The key challenges in single molecule tracking (SMT) studies in live cells remain the assignment of biological meaning from a set of molecule trajectories, as well as the technical challenges in generating the volume of data necessary to characterize interaction pathways. Through advancements in optical engineering, automation, and computation, we have developed an industrial-scale system to track fast-moving proteins across thousands of conditions in mammalian cell lines. We applied this system to study the estrogen receptor (ER), a protein well known for its role in normal human development as well as its prominent role in many breast cancers. We present a chemical genetics screen of thousands of known bioactive compounds to identify novel ER modulators. Intriguingly, chemical genetics also revealed the contributions of multiple pathway interactions on ER dynamics, distinguishable through pathway-specific effects. Further, SMT can be an effective readout for direct target engagement via real-time kinetics measurements, can elucidate structure-activity relationships, and can predict compound efficacy in orthogonal assays. Taken together, our results underscore the wealth of information embedded within high-throughput, live-cell SMT data, and the utility of this information in furthering our understanding of biology across a broad set of applications.

Background

Protein interaction networks are key to biology but hard to quantify

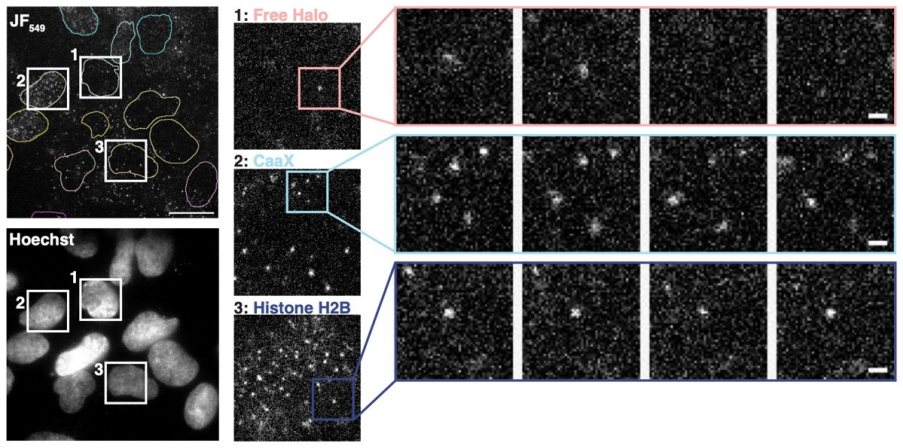
JF549
Hoechst
1: Free Halo
2: CaaX
3: Histone H2B

- Single-molecule-resolution measurements in live cells, in near real-time, provide a means to quantify interaction networks[1]
- Changes in interactions can manifest as changes in protein dynamics
- Challenging to scale to collect enough data to dissect interaction networks, rather than just binary interactions

Estrogen Receptor (ER) is an example of how an interaction network drives biology

- Mis-regulation of ER is the primary driver in breast cancer[2]
- Regulation of ER involves changes in its binding partners: under basal conditions, ER is sequestered in complex with HSP90 and other cofactors. Upon ligand binding, the receptor dissociates from the inactive complex, dimerizes, and binds to DNA.[3]

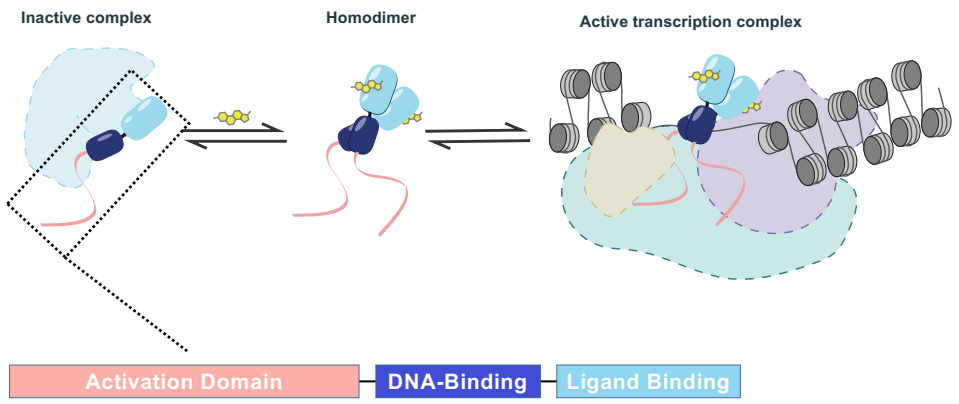
Inactive complex
Homodimer
Active transcription complex
Activation Domain
DNA-Binding
Ligand Binding

High-throughput SMT detects ER modulators

We have created a high-throughput SMT platform to measure protein dynamics in live cells

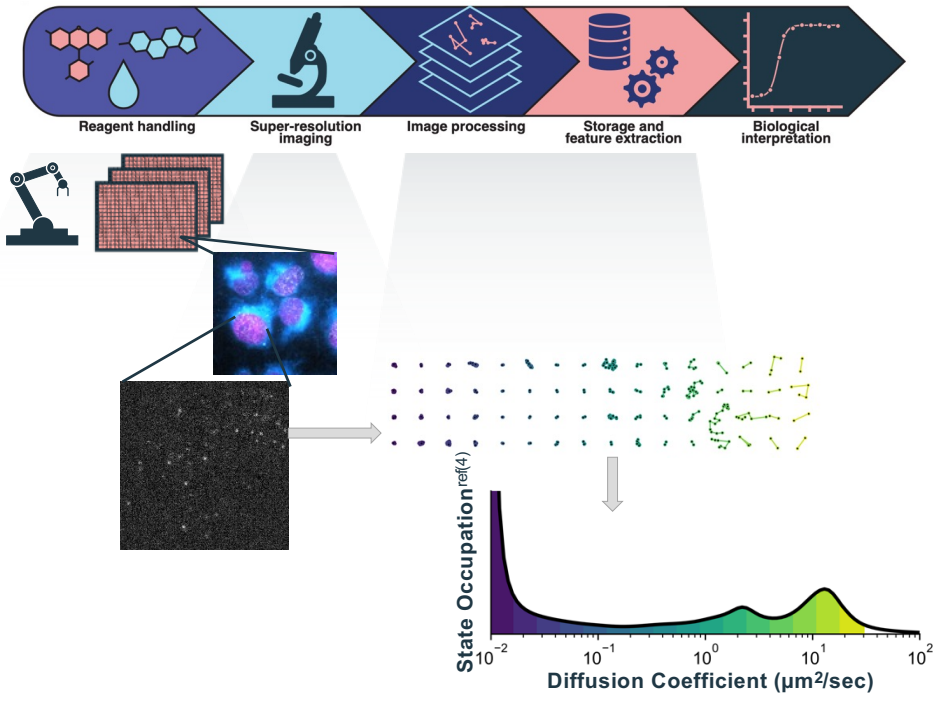
Reagent handling
Super-resolution imaging
Image processing
Storage and feature extraction
Biological interpretation
State Occupation ref(4)
Diffusion Coefficient (μm²/sec)

Small-molecule effects on ER are detected as changes to distributions of diffusive states

- Estradiol promotes binding of ER to chromatin[5]
- Observed in htSMT as an increase in "fraction bound"

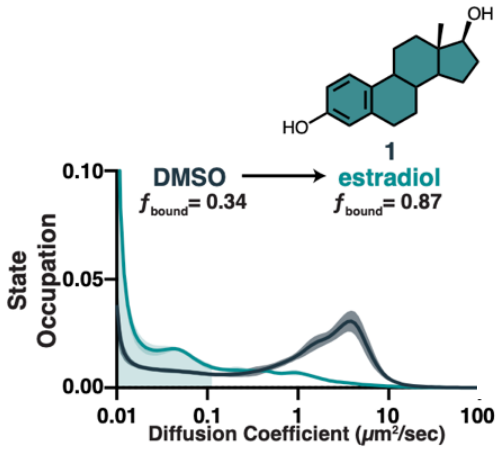
OH
HO
1
DMSO
f bound = 0.34
estradiol
f bound = 0.87
State Occupation
Diffusion Coefficient (μm²/sec)

Bioactive compound screen identifies known and novel modulators of ER

- We screened a structurally diverse set of 5,067 known bioactive compounds and measured their effects on the bound fraction of ER
- Known direct ER modulators ("ER positive controls") increased the bound fraction
- Known pathway modulators increased or decreased the bound fraction
- 209 new ER modulators were identified

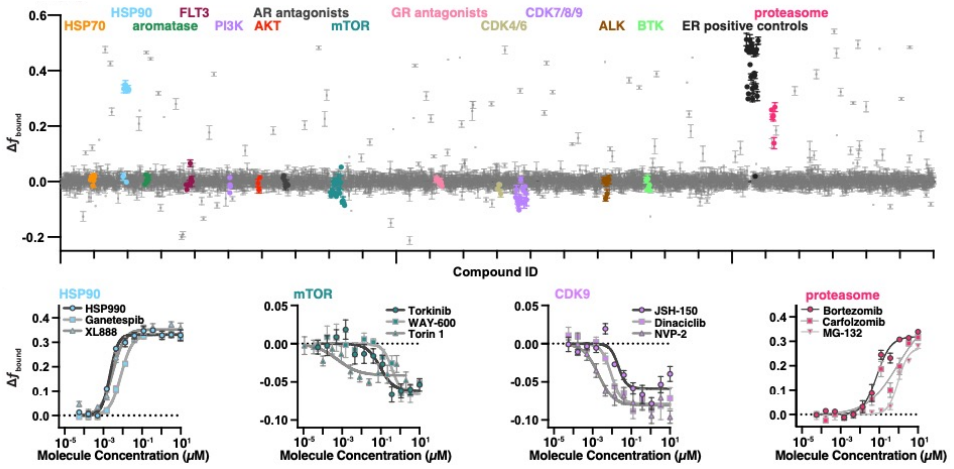
HSP70
HSP90
aromatase
FLT3
PI3K
AKT
AR antagonists
mTOR
GR antagonists
CDK4/6
CDK7/8/9
ALK
BTK
ER positive controls
proteasome
Compound ID
Δf bound
HSP990
Ganetespib
XL888
Torkinib
WAY-600
Torin 1
CDK9
JSH-150
Dinaciclib
NVP-2
Bortezomib
Carfolzomib
MG-132
Molecule Concentration (μM)

Direct ER modulators can be distinguished from pathway perturbations by time to activity

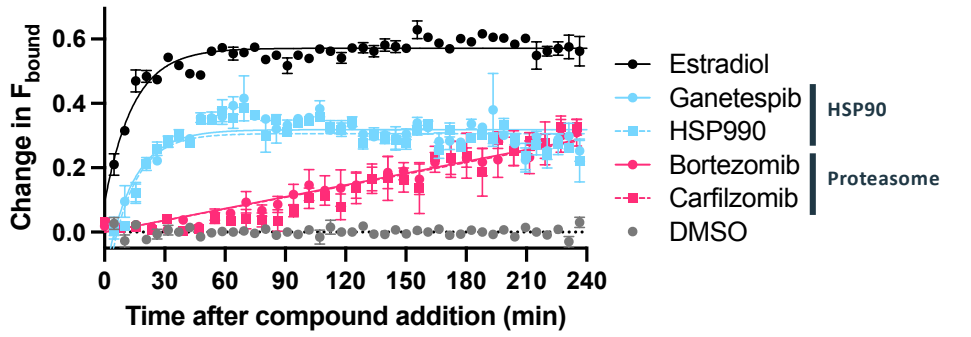
Change in F bound
Time after compound addition (min)
Estradiol
Ganetespib
HSP990
HSP90
Bortezomib
Carfilzomib
Proteasome
DMSO

- We can measure fraction bound at different times post compound addition
- Direct ER modulators affect fraction bound almost immediately; pathway perturbations take longer to show an effect
- We can order these effectors by how long they take to change fraction bound, enabling us to begin building an interaction network model:

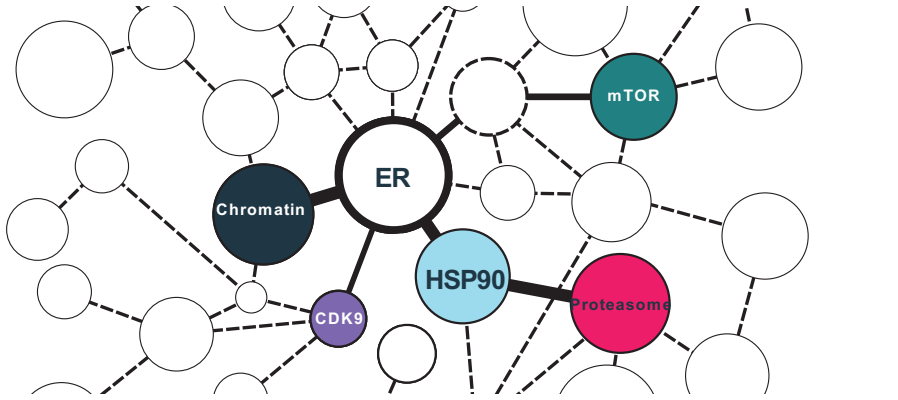
ER
Chromatin
mTOR
HSP90
CDK9
Proteasome

SMT differentiates between steroid hormone receptors and their cognate hormones

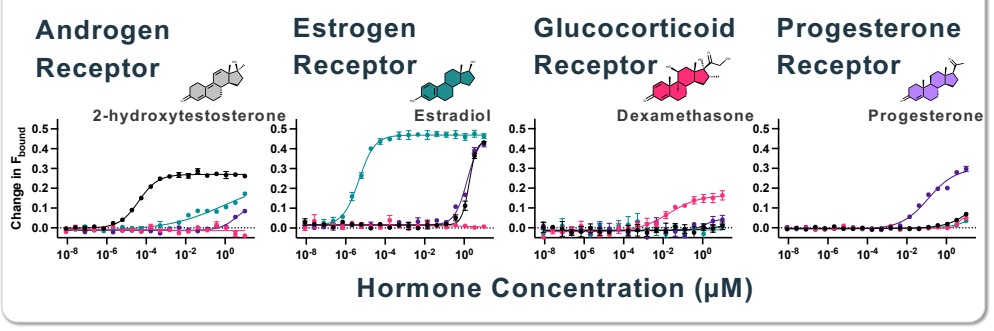
Androgen Receptor
Estrogen Receptor
Glucocorticoid Receptor
Progesterone Receptor
2-hydroxytestosterone
Estradiol
Dexamethasone
Progesterone
Change in F bound
Hormone Concentration (μM)

Conclusions

- We have developed an automated platform capable of performing SMT on more than one million cells per day and quantifying changes in diffusive states after compound addition
- Using this platform, we can identify all 30 known steroid ligands from a library of 5,067 bioactive compounds, plus 209 non-steroidal molecules
- By interrogating the change in ER dynamics as a function of time after compound addition, we can model a network of ER interactors in living cells

Acknowledgements

We thank the Eikon Therapeutics Co-Founders and Scientific advisors for their expertise and advice. We also thank the entire Eikon team for their supporting work.

References

1) Shen H, Tauzin LJ, Baiyasi R, Wang W, Moringo N, Shuang B, Landes CF. *Chem Rev*, **2017**, 117, 7331-7376.
2) Lu Y, Liu W. *J Med Chem,* **2020,** 63 (24), 15094-15114.
3) Saha S, Dey S, Nath S. *Front Oncol*, **2021**, 11, 620214.
4) Heckert A, Dahal L, Tijan R, Darzacq X. *Elife*, **2022**, 11.
5) Paakinaho V, Presman DM, Ball DA, Johnson TA, Schiltz RL, Levitt P, Mazza D, Morisaki T, Karpova TS, Hager GL. *Nat Comm,* **2017**, 8, 15896.