Characterizing primary biliary cholangitis (PBC) patients in the real world: demographics and treatment characteristics of the PicnicHealth PBC cohort

Poster No. 4775

Usha Gungabissoon[1], Helen T Smith[1]*, Robyn von Maltzahn[1], John Logie[1], Jolyon Fairburn-Beech[1], Liyuan Ma[2], Sivangi Nair[3], Dhirishiya P[3], Ashleigh McGirr[4], Haley S Friedler[5], Meghan Tierney[5], Christopher Rowe[5]

[1]GSK, London, UK; [2]GSK, Collegeville, PA, USA; [3]GSK, Bangalore, India; [4]GSK, Mississauga, ON, Canada; [5]PicnicHealth, San Francisco, CA, USA

*Presenting author

Background

- Up to 74% of patients with primary biliary cholangitis (PBC), a rare disease, will be affected by cholestatic pruritus over the course of their illness.[1–4]
- Cholestatic pruritus significantly impairs quality of life (QoL) and is associated with a negative impact on physical and emotional well-being that increases with pruritus severity.[5,6]
- Administrative healthcare databases (AHD), used routinely by medical providers to record clinical and laboratory data, are widely used to provide insight into the comorbidity burden and treatment use of patients in real-world settings.
- Whilst management of PBC in clinical practice is recorded and can thus be described using these datasets, symptoms, such as pruritus, are not.[7]
 - The off-label use of medications to manage pruritus (that are licensed for non-pruritus indications), as well as the absence of a specific International Classification of Diseases (ICD) code for cholestatic pruritus, makes it difficult to reliably describe management of itch using data from AHD.
 - Additionally, there are no means of evaluating the severity or impact of pruritus and other symptoms of PBC due to lack of health-related quality of life (HRQoL) measures in AHD.
- To overcome these limitations and understand the severity and prevalence of pruritus, PicnicHealth has compiled a real-world, de-identified, patient-level database of structured and narrative data from medical records of patients with PBC.[8]
- These medical record data are supplemented by the PBC-40, a disease-specific patient-reported outcome (PRO) measure used to evaluate the impact of PBC on patients' QoL[9], including the severity of itch.

Aim

Here, we characterize patients in the PicnicHealth PBC cohort to assess the representativeness of the database and its utility for further studies.

Methods

Study design, recruitment and data collection are summarized in **Figure 1**.

The analysis population included all patients who had completed the PBC-40 itch domain at baseline (defined as enrolment into the cohort).

Descriptive statistics summarized patient demographics and PBC-related clinical measures, including PBC staging data, which was abstracted directly from patients' medical records where available.

Figure 1. Study design

Recruitment

Since **Jan 2021**

Patients with a **confirmed diagnosis of PBC** in the USA

Recruited by PicnicHealth via:
- Patient advocacy groups
- Affiliate partners
- Social media campaigns

Consented to provide medical records for research

Data Collection

Collection of patient's entire medical record*

Structured data: Condition | Medication | Vitals | Procedure | Labs | Genetics

Unstructured data: Narrative text e.g. doctor's notes

Machine learning platform parses and extracts data, human annotators verify integrity → Data extracted from **narrative text** → **Clinical concepts extracted** and mapped to standard ontologies† → De-identified **patient-level database** generated

~7–8 years retrospective data and prospectively collected records

PBC-40 questionnaire
- **Disease-specific HRQoL measure** comprising 40 questions‡ grouped into six domains
- Issued at **baseline and quarterly** thereafter

Data Analysis

Patient demographics and baseline clinical characteristics including:
- Duration of PBC
- Age at diagnosis
- PBC stage
- Baseline ALP
- Recent PBC treatment§

Baseline PBC-40 score for all domains:
- Symptoms
- Itch
- Fatigue
- Cognition
- Social
- Emotional

*Medical records are received in any format used by the transmitting facility or provider. PDF and paper documents are processed by an optical character recognition system to generate searchable text from the original images. †Systematized Nomenclature of Medicine (SNOMED), International Classification of Diseases (ICD), Logical Observation Identifiers Names and Codes (LOINC), RxNorm. ‡Questions are scored on a scale of 0 or 1 to 5, where 0 = not present, 1 = least impact, 5 = greatest impact. §Defined as documentation in the patients' structured and unstructured medical record of treatment with ursodeoxycholic acid (UDCA), obeticholic acid, or fibrates in the 12 months prior to baseline. ALP, alkaline phosphatase.

Results

Baseline demographics and clinical characteristics

As of May 2022, 225 patients were recruited and included in the analysis. Baseline demographics, clinical characteristics and treatment use are shown in **Figure 2** and **Table 1**.

Figure 2. Key baseline characteristics for the analysis population*

- Sex: **98% (n=220)** female
- Mean (SD) age: **55.4 (10.9)** years†
- Mean (SD) duration of PBC‡: **5.1 (5.5)** years
- Mean (SD) age at PBC diagnosis: **50.3 (10.4)** years

*All patients who had completed the PBC-40 itch domain. †At baseline. ‡Time since first recorded PBC diagnosis to baseline was used as a proxy for PBC duration.
SD, standard deviation.

Table 1. Clinico-demographic characteristics for the analysis population*

	PicnicHealth PBC cohort N=225
Ethnicity, n (%)	
Not Hispanic or Latino	204 (91)
Hispanic or Latino	16 (7)
Prefer not to say	5 (2)
Baseline ALP†, mean (SD), IU/L	188.1 (141.0)
Cirrhosis, n (%)	66 (29)
Ever‡ PBC treatment, n (%)	
UDCA	219 (97)
Obeticholic acid	46 (20)
Fibrates	19 (8)
Recent§ PBC treatment, n (%)	
UDCA	196 (87)
Obeticholic acid	37 (16)
Fibrates	17 (8)
Recent§ anti-pruritic treatment¶, n (%)	120 (53)
Bile acid sequestrant	29 (13)
Sertraline	31 (14)
Rifampicin	7 (3)
Naltrexone/Nalmefene	13 (6)
Antihistamines	96 (43)
Other anti-pruritics#	5 (2)

*All patients who had completed the PBC-40 itch domain. †Closest to baseline but within 12 months. ‡Ever reported in medical record, based on RxNorm. §Within 12 months prior to baseline, based on RxNorm. ¶Indication for use may or may not have been pruritus. #Includes ondansetron, metronidazole or propofol.

PBC staging data was available for a subset of patients (n=82; **Figure 3**).

Figure 3. PBC disease stage for patients included in the PicnicHealth PBC cohort*

PBC stage (n=82)

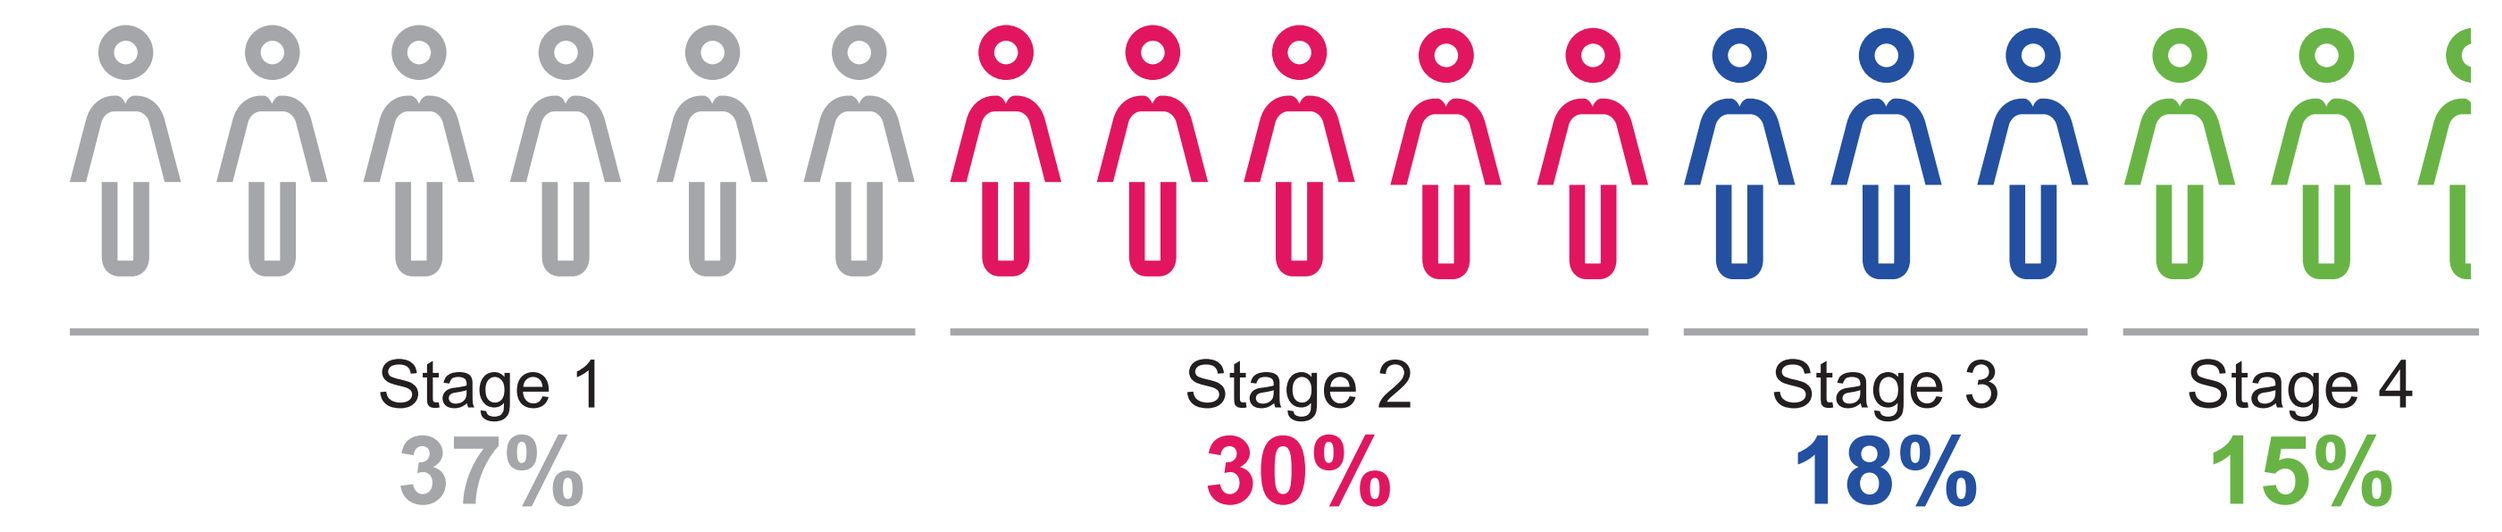
Stage 1
37%
Stage 2
30%
Stage 3
18%
Stage 4
15%

Each icon represents five patients. Numbers sum to >100% due to rounding. *Where available, staging data were extracted directly from medical records.

PBC-40 baseline data

Mean scores for each domain of the PBC-40 in the entire cohort are shown in **Figure 4**. In the itch domain, 89% (n=200) patients reported itch of any severity (score ≥1).

Figure 4. PBC-40 domain scores at baseline

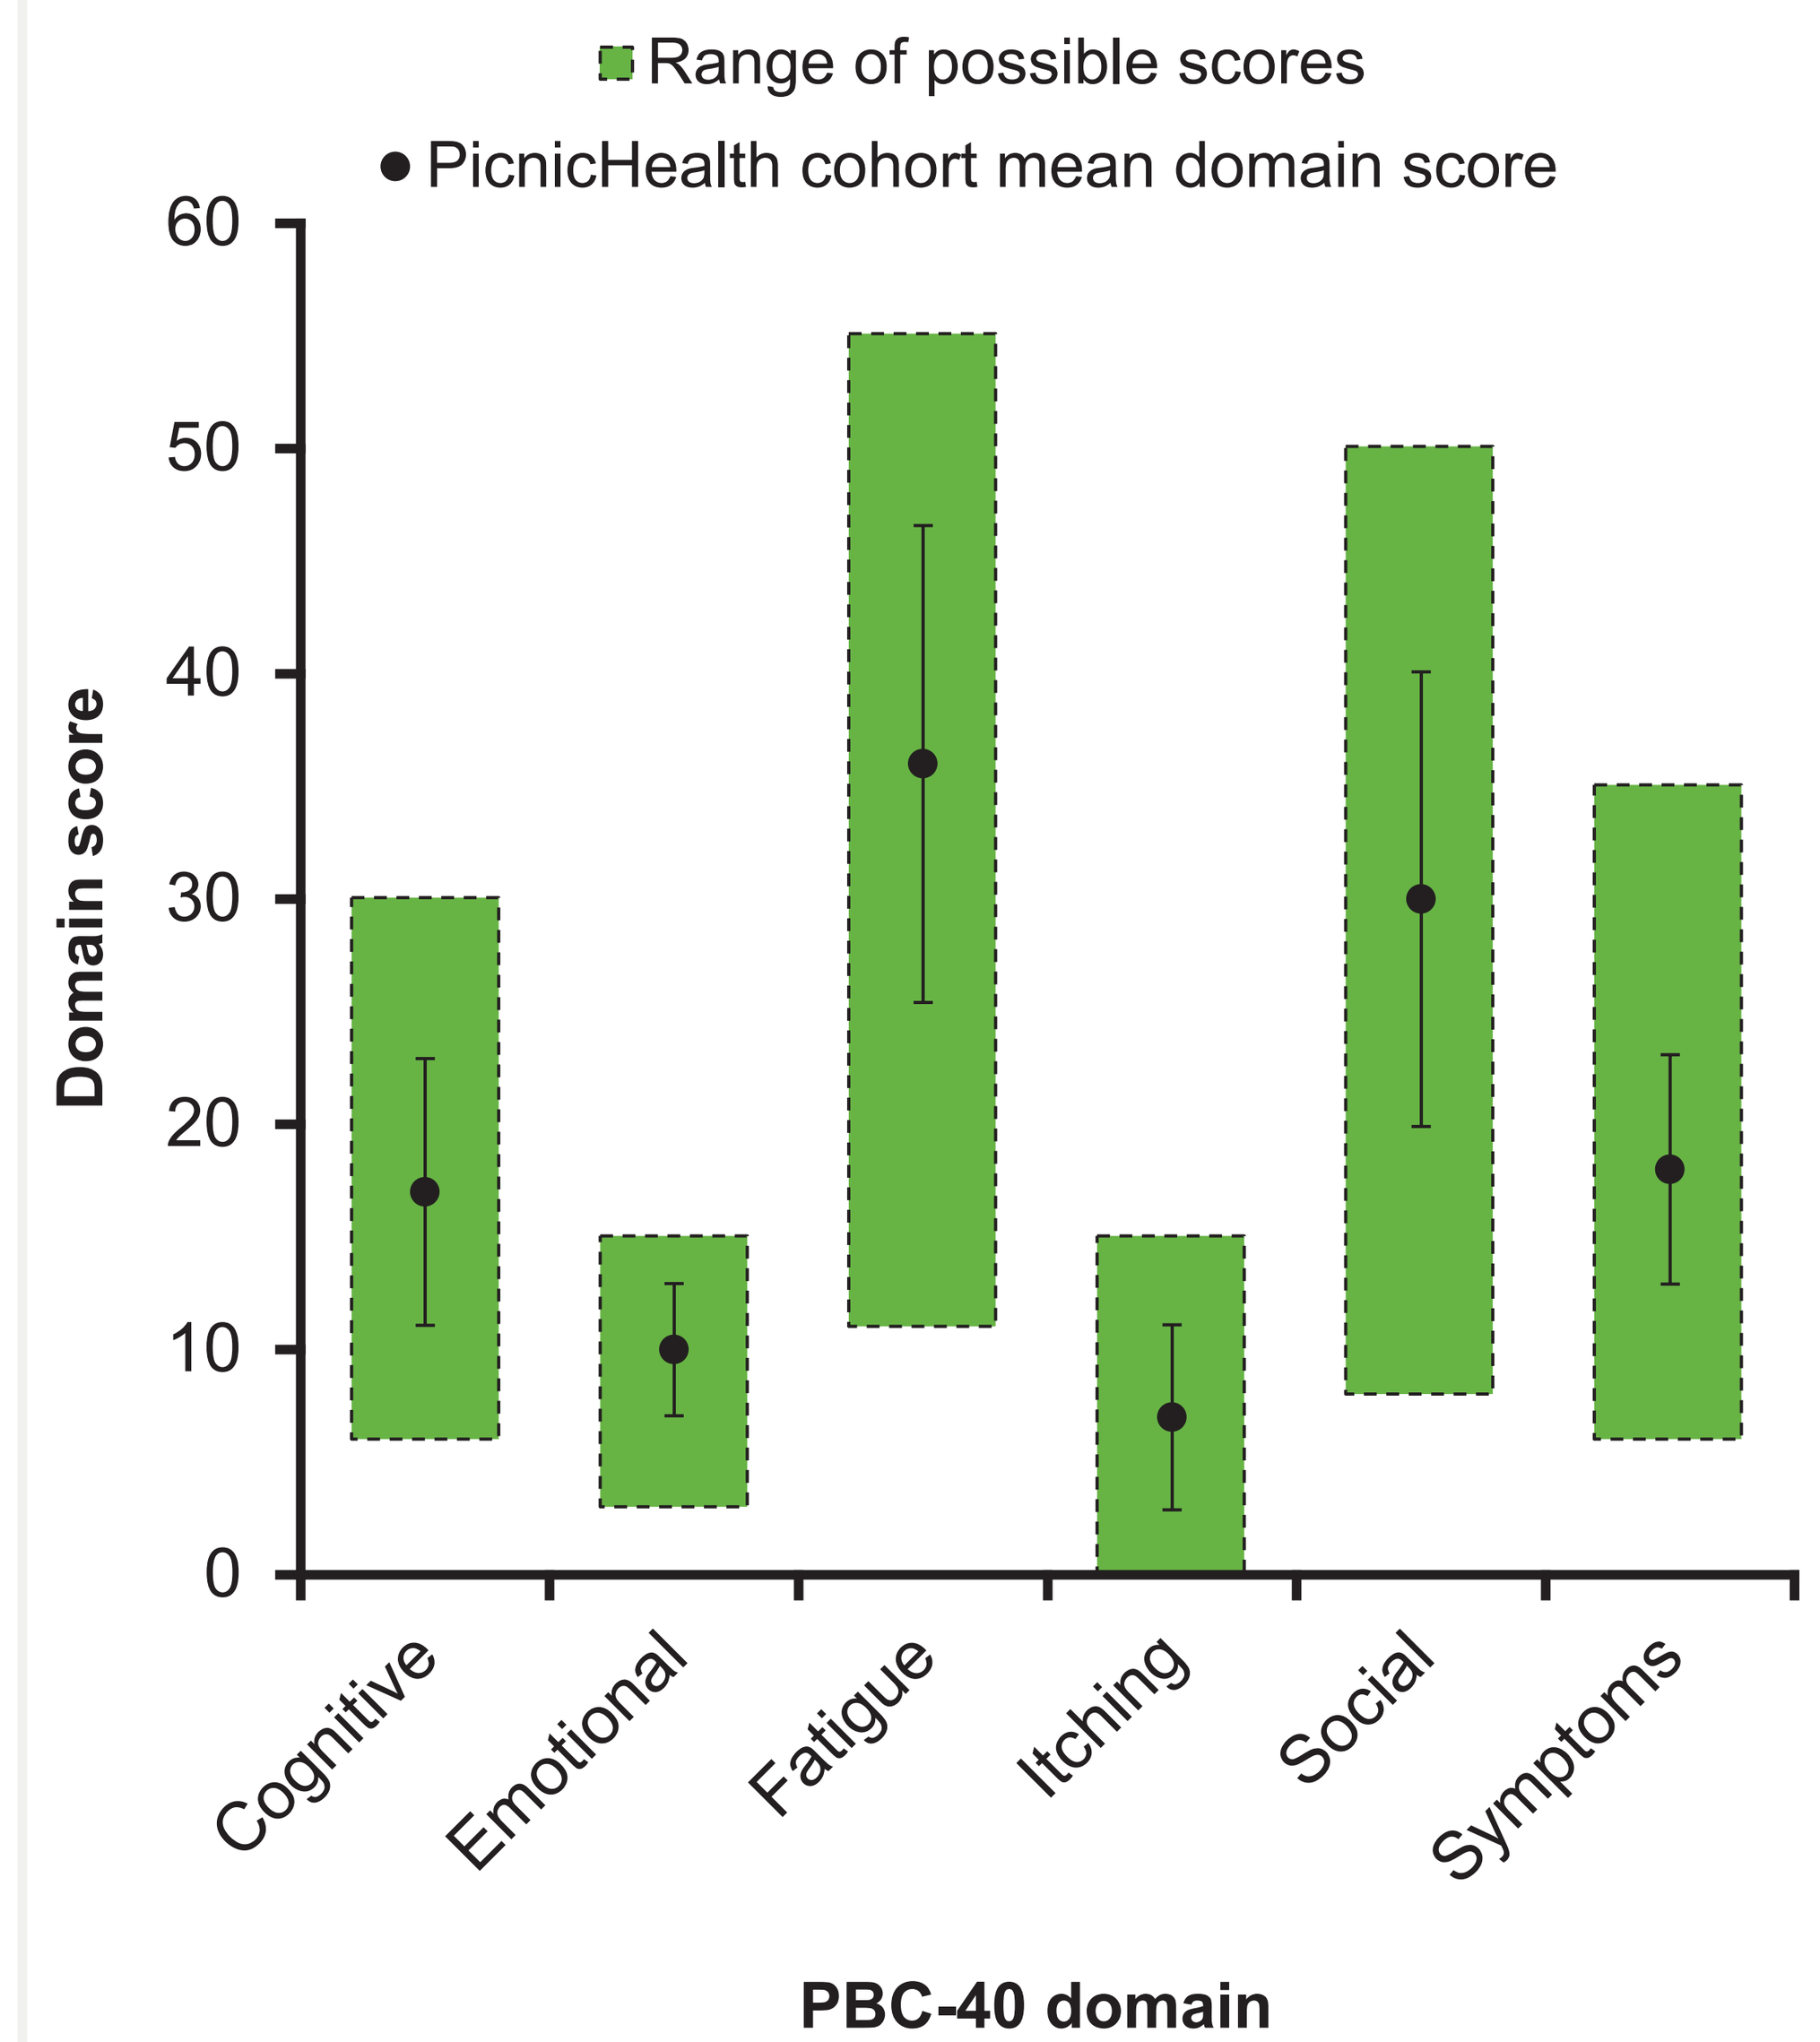
Range of possible scores
PicnicHealth cohort mean domain score
Domain score
60
50
40
30
20
10
0
Cognitive
Emotional
Fatigue
Itching
Social
Symptoms
PBC-40 domain

Error bars represent SD. Green bars represent possible range of values for each domain.
N values: Cognitive, Fatigue, Itching and Symptoms = 225, Emotional and Social = 223.

Conclusions

The clinico-demographic profile of the PicnicHealth PBC cohort is broadly consistent with the expected profile of patients with PBC; however, there was a higher proportion of female participants and a higher proportion of patients reporting pruritus than expected.

- One possible explanation for this is selection bias; patients with severe PBC-related pruritus may be more likely to enroll in the study as it offers the opportunity to access their entire medical history in a convenient format.

Mean scores for each domain of the PBC-40 were generally consistent with those reported in the literature, including the recent analysis of the TARGET-PBC cohort.[6,9,10]

As the PicnicHealth database includes PROs, it could provide a future resource for further characterization of patients with PBC and pruritus.

Further analysis of overlap of pruritus recorded in medical records with patient-reported pruritus is reported in **Poster 4748**.

Disclosures

UG, **HTS**, **RvM**, **JL**, **JF-B**, **LM**, **SN**, **DP**, and **AM** are employees of, and hold shares in GSK.

HSF, **MT**, and **CR** are employees of, and hold stock options in PicnicHealth.

Study conduct and data analysis were performed by PicnicHealth funded by GSK. No funding was provided to PicnicHealth for poster development.

Ashleigh McGirr was added as a poster author and contributed to poster development, but was not included on the original abstract. All authors were informed of, and agreed to, her inclusion as a poster author.

Acknowledgments

This study was funded by GSK (Study 218574).

Editorial support (in the form of in the form of writing poster content, including development of the initial draft based on author direction, incorporating author comments, developing figures, layout assistance, and grammatical editing) was provided by Sheekha Amin, MPharmacol, of Fishawack Indicia Ltd, UK, part of Fishawack Health, and was funded by GSK.

References

1. Lindor KD, et al. *Hepatology* 2019;69:394–419.
2. Trivedi HD, et al. *Am J Med* 2017;130:744.e1–7.
3. Hegade VS, et al. *Clin Gastroenterol Hepatol* 2019;17:1379–87.
4. Talwaklar JA, et al. *Clin Gastroenterol Hepatol* 2003;1:297–302.
5. Jin XY, Khan TM. *J Formos Med Assoc* 2016;115:689–702.
6. Mayo MJ, et al. *Dig Dis Sci* 2022;1–11.
7. Gungabissoon U, et al. *BMJ Open Gastroenterol* 2022;9(1):e000857.
8. PicnicHealth. https://assets.website-files.com/6074d822c3c38b07ef63efe5/608abcf69bafeb1b2a49b16e_Why%20PicnicHealth%20Data.pdf [Accessed August 2022].
9. Jacoby A, et al. *Gut* 2005;54:1622–9.
10. Blackburn P, et al. *Liver Int* 2007;27(5): 654–61.

Please scan the QR code to access the poster

Please scan the QR code to access the related **Poster 4748** presented at this meeting.

Author email address: helen.t.smith@gsk.com

GSK

Presented at the American Association for the Study of Liver Disease (AASLD) The Liver Meeting, Washington DC, USA, November 4–8, 2022